Bimekizumab versus Secukinumab for the Treatment of Nail Psoriasis in Patients with Moderate to Severe Plaque Psoriasis: Results from the BE RADIANT Phase 3b Trial

2255

Kilian Eyerich,[1] Alice B. Gottlieb,[2] Stefano Piaserico,[3] Stefan Beissert,[4] Melinda Gooderham,[5] Brian Kirby,[6] Nicola Tilt,[7] Cynthia Madden,[8] Susanne Wiegratz,[9] Dirk de Cuyper,[10] Joseph F. Merola,[11] Boni Elewski[12]

Objective

To compare the efficacy of bimekizumab (BKZ) versus secukinumab (SEC) through 48 weeks for the treatment of nail psoriasis, including complete nail clearance, in patients with moderate to severe plaque psoriasis who had nail involvement at baseline.

To compare modified Nail Psoriasis Severity Index (mNAPSI) sub-score complete clearance rates at Week 16 and Week 48 in patients treated with BKZ versus SEC.

Background

- Psoriatic lesions in highly visible areas, including the nails, disproportionately affect patients' health-related quality of life.[1]
- Here, we assessed mNAPSI total scores and mNAPSI sub-scores among patients with baseline mNAPSI >10 in the BE RADIANT phase 3b trial.[2]

Methods

- In BE RADIANT, patients were randomized to BKZ 320 mg every 4 weeks (Q4W) or SEC 300 mg weekly to Week 4 then Q4W.[2] From Week 16, BKZ-randomized patients received BKZ Q4W or Q8W (**Figure 1**).
- Modified Nail Psoriasis Severity Index (mNAPSI) scores (total and sub-scores) are reported for BKZ- and SEC-randomized patients who had baseline mNAPSI >10 and entered the maintenance period (**Table 1**).
- Results for BKZ-treated patients are reported as BKZ Total, comprising all patients who received ≥1 BKZ dose in the maintenance period (combined Q4W/Q4W and Q4W/Q8W groups).
- Missing data were handled using non-responder imputation (NRI). Modified non-responder imputation (mNRI) and observed cases (OC) are also reported.

Results

- In total, 116 BKZ-treated and 99 SEC-treated patients had baseline mNAPSI >10 and entered the maintenance period (**Table 1**); 37.1% and 40.4% of BKZ- and SEC-treated patients achieved mNAPSI ≤2 at Week 16, respectively; 75.0% and 59.6% achieved mNAPSI ≤2 at Week 48 (**Figure 2**).
- From Weeks 16–48, mNAPSI 0 (clear) response rates increased from 26.7% to 70.7% with BKZ, and 33.3% to 53.5% with SEC.
- At Week 16, 36.6% of BKZ-treated and 41.9% of SEC-treated patients achieved a nail pitting sub-score of 0 (clear), increasing to 78.2% and 59.3% of patients at Week 48 (**Figure 3**).
- Of BKZ-treated patients at Week 16, 65.2% achieved a nail plate crumbling sub-score of 0 (clear), vs 69.6% of SEC-treated patients. Achievement increased to 85.5% and 75.0% at Week 48, respectively (**Figure 3**).
- Of BKZ-treated patients at Week 16, 69.5% achieved a leukonychia sub-score of 0 (clear), vs 70.7% of SEC-treated patients. Achievement increased to 79.7% and 69.0% of patients at Week 48 (**Figure 3**).

Conclusions

Complete nail clearance (mNAPSI 0) was achieved in numerically higher proportions of bimekizumab-treated patients than secukinumab-treated patients by Week 48.

At Week 48, bimekizumab-treated patients showed numerically higher rates of complete clearance for all mNAPSI sub-scores compared with secukinumab-treated patients, with the exception of red spots in lunula.

Summary

mNAPSI[a]

Each nail is scored from 0–13 as follows:

0–3 for:

- Onycholysis/oil-drop dyschromia
- Nail plate crumbling
- Pitting

0 (no) or 1 (yes) for:

- Leukonychia
- Nail bed hyperkeratosis
- Splinter hemorrhages
- Red spots in lunula

mNAPSI is the sum of the scores for all nails (range: 0–130, 0 indicates clear nails)

Here, we assessed mNAPSI total scores and mNAPSI sub-scores among patients with baseline mNAPSI >10

Complete nail clearance (mNAPSI 0) was achieved in numerically higher proportions of BKZ-treated patients than SEC-treated patients by Week 48

[a] The mNAPSI has demonstrated superior interrater variability and correlations with patient and physician global assessments versus the NAPSI.[3]

Table 1 Baseline characteristics: included patients

	BKZ Total[a] N=116	SEC N=99
Age (years), mean ± SD	45.7 ± 14.0	43.9 ± 12.8
Male, n (%)	94 (81.0)	81 (81.8)
White, n (%)	109 (94.0)	97 (98.0)
Weight (kg), mean ± SD	93.0 ± 21.0	90.5 ± 17.9
Duration of psoriasis (years), mean ± SD	20.1 ± 14.3	17.3 ± 10.6
PASI, mean ± SD	21.8 ± 8.9	20.4 ± 6.3
BSA (%), mean ± SD	27.4 ± 18.3	23.4 ± 12.7
IGA, n (%)		
3: moderate	60 (51.7)	71 (71.7)
4: severe	55 (47.4)	28 (28.3)
DLQI total score, mean ± SD	10.6 ± 6.7	12.3 ± 7.5
mNAPSI, mean ± SD	28.5 ± 17.9	29.6 ± 20.6
Onycholysis/oil-drop dyschromia	9.0 ± 7.5	9.1 ± 7.3
Nail plate crumbling	4.4 ± 6.4	4.1 ± 6.0
Pitting	8.4 ± 7.0	8.4 ± 7.3
Leukonychia	2.0 ± 2.9	2.9 ± 3.6
Nail bed hyperkeratosis	2.4 ± 3.2	2.8 ± 3.8
Splinter hemorrhages	1.6 ± 2.5	1.5 ± 2.5
Red spots in lunula	0.6 ± 1.7	0.7 ± 2.2
Any prior systemic therapy, n (%)	87 (75.0)	79 (79.8)
Any prior biologic therapy, n (%)	45 (38.8)	42 (42.4)

[a] Data were pooled for all patients who received ≥1 BKZ dose at Week 16 or later (BKZ Total). BKZ Total included all patients with mNAPSI>10 at baseline who were treated with BKZ Q4W/Q4W (n=42) or with BKZ Q4W/Q8W (n=74), combined.

Figure 1 BE RADIANT study design

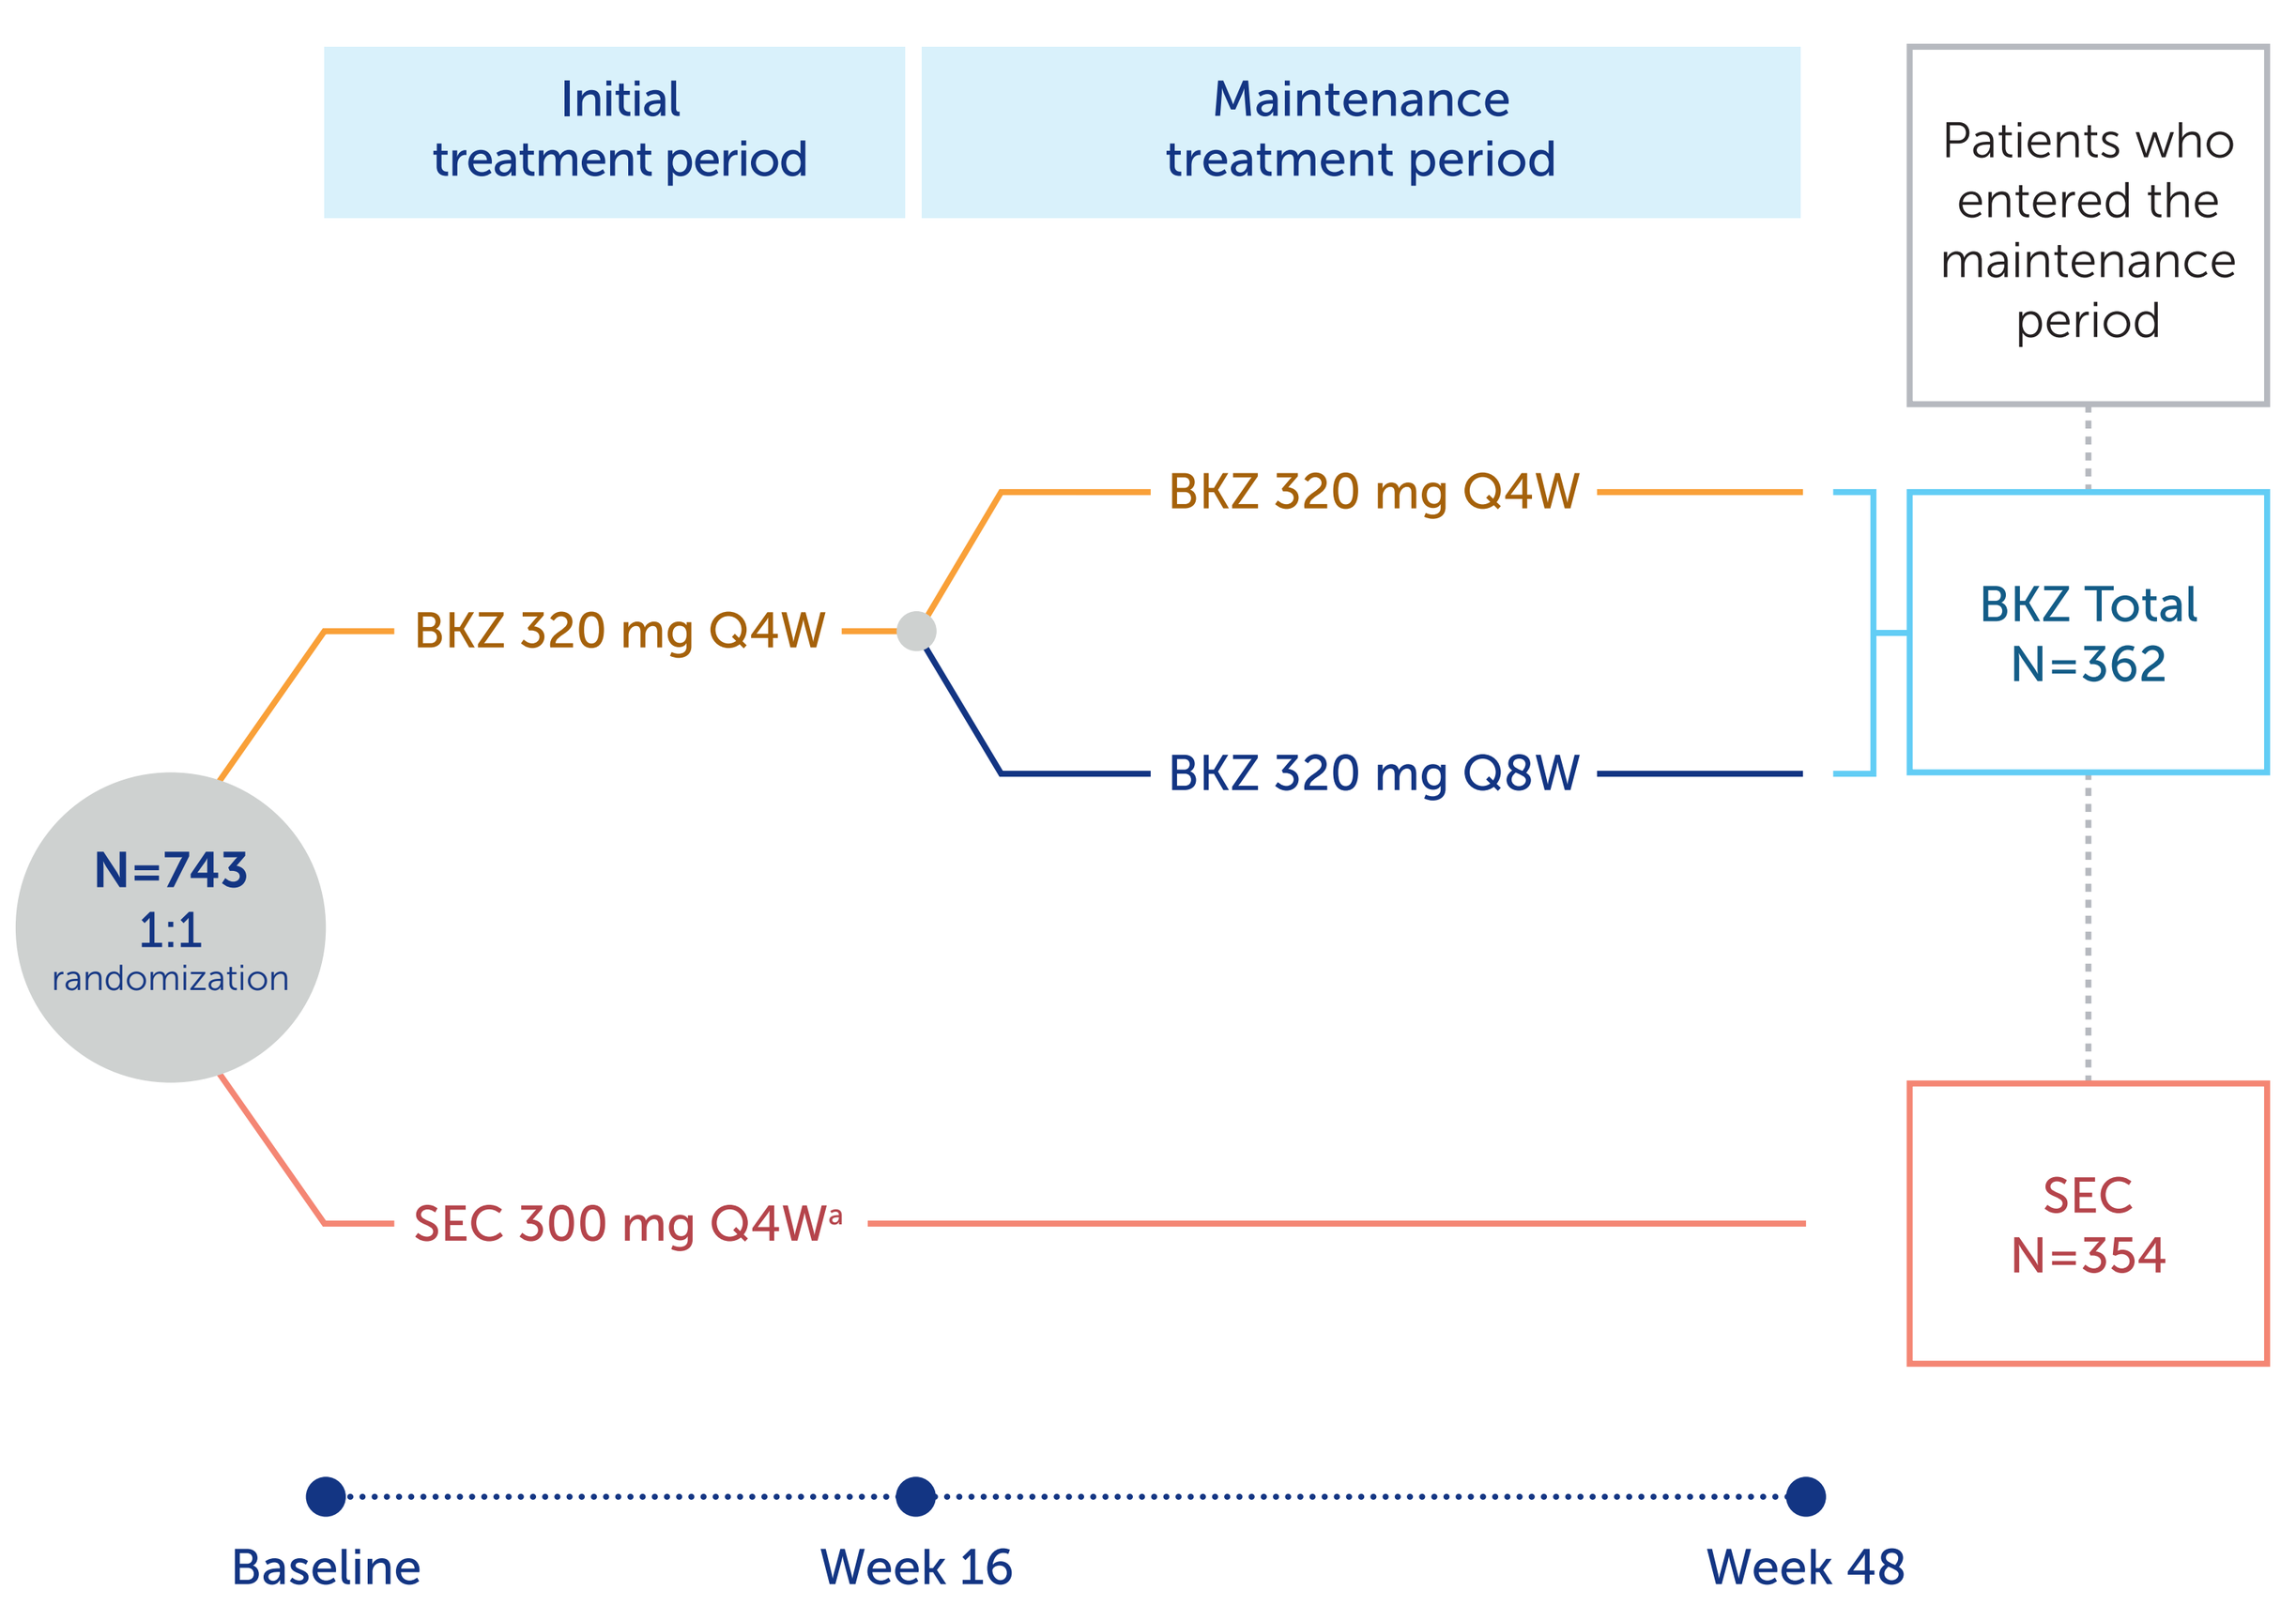
Initial treatment period
Maintenance treatment period
Patients who entered the maintenance period
N=743
1:1
randomization
BKZ 320 mg Q4W
BKZ 320 mg Q4W
BKZ 320 mg Q8W
SEC 300 mg Q4W[a]
BKZ Total N=362
SEC N=354
Baseline
Week 16
Week 48

[a] SEC was dosed 300 mg weekly until Week 4, then Q4W for the remainder of the double-blinded treatment period.

Figure 2 mNAPSI ≤2 and mNAPSI 0 through Week 48 in patients with baseline mNAPSI >10 (NRI)

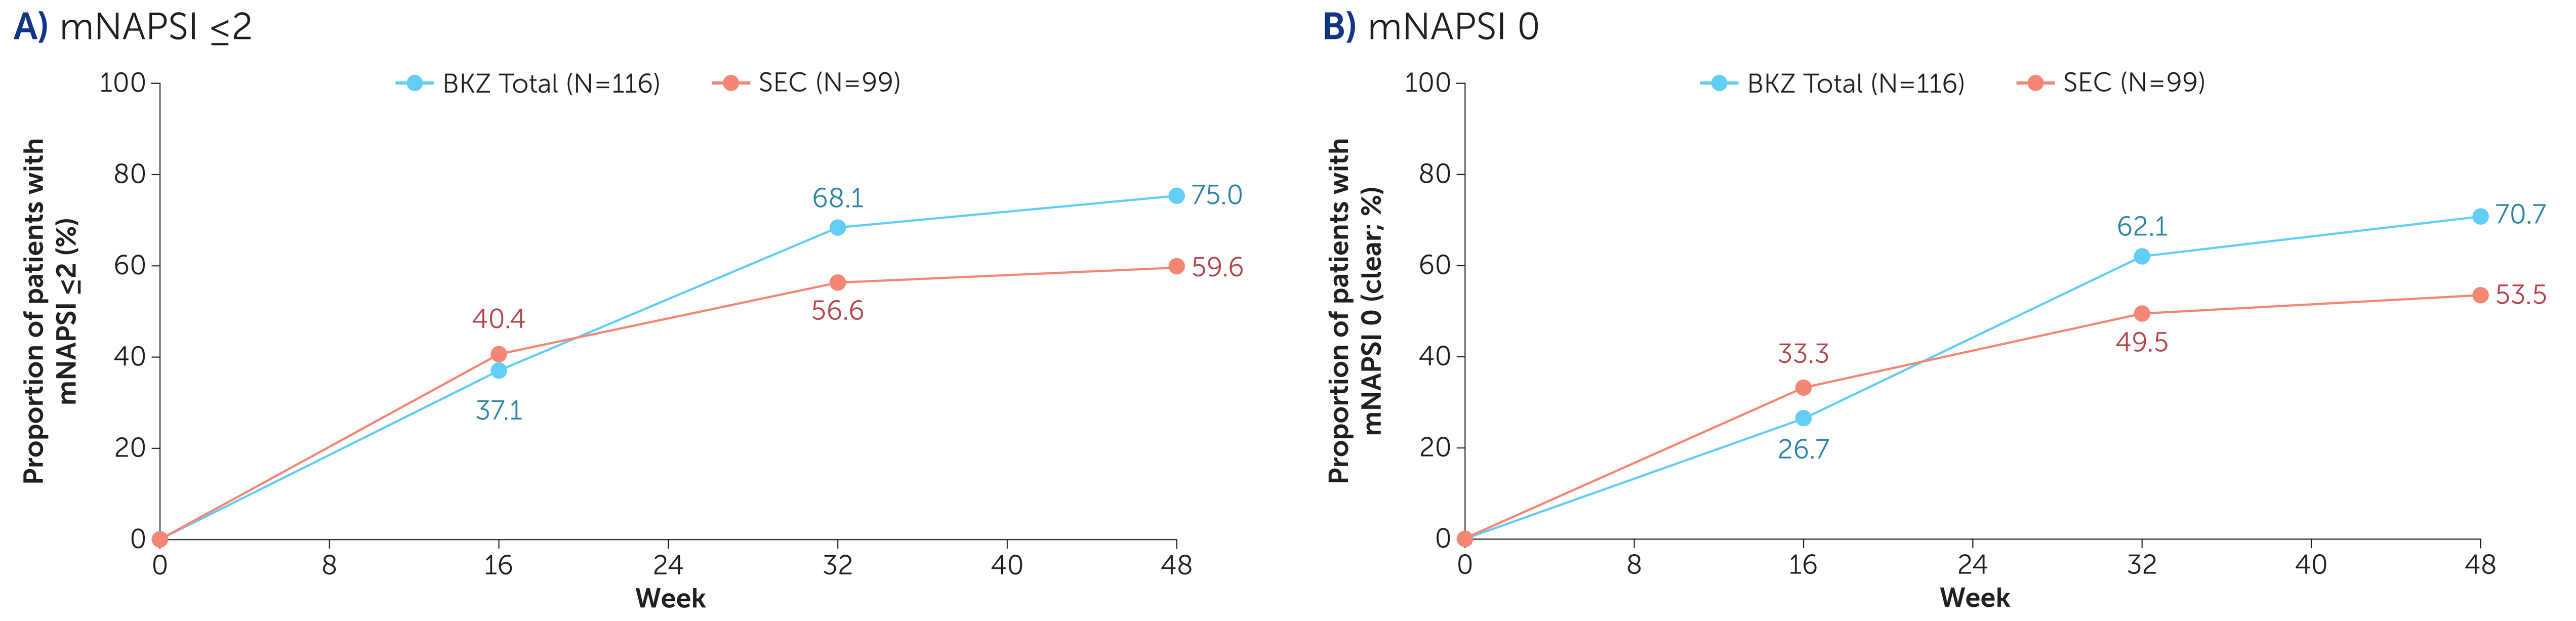
A) mNAPSI ≤2
BKZ Total (N=116)
SEC (N=99)
Proportion of patients with mNAPSI ≤2 (%)
40.4
37.1
68.1
56.6
75.0
59.6
Week
B) mNAPSI 0
BKZ Total (N=116)
SEC (N=99)
Proportion of patients with mNAPSI 0 (clear; %)
33.3
26.7
62.1
49.5
70.7
53.5
Week

A) mNAPSI ≤2

Week		16	32	48
OC; % (n/N)	BKZ Total	37.7 (43/114)	70.5 (79/112)	81.3 (87/107)
	SEC	40.4 (40/99)	60.9 (56/92)	67.8 (59/87)
mNRI;[a] %	BKZ Total	37.6	70.3	78.6
	SEC	40.4	59.3	66.2
NRI; %	BKZ Total	37.1	68.1	75.0
	SEC	40.4	56.6	59.6

B) mNAPSI 0

Week		16	32	48
OC; % (n/N)	BKZ Total	27.2 (31/114)	64.3 (72/112)	76.6 (82/107)
	SEC	33.3 (33/99)	53.3 (49/92)	60.9 (53/87)
mNRI;[a] %	BKZ Total	27.1	63.5	73.5
	SEC	33.3	51.5	58.1
NRI; %	BKZ Total	26.7	62.1	70.7
	SEC	33.3	49.5	53.5

Only patients with mNAPSI >10 at baseline and who entered the maintenance period of BE RADIANT were included in this analysis. A score of 0 indicates clear nails. [a] For mNRI, patients who discontinued study treatment due to lack of efficacy or adverse events deemed treatment-related by investigators were considered non-responders at subsequent timepoints; multiple imputation was used for all other missing data.

Figure 3 Complete clearance of mNAPSI sub-scores at Week 16 and Week 48 in patients with baseline mNAPSI >10 and sub-score >0 (NRI)

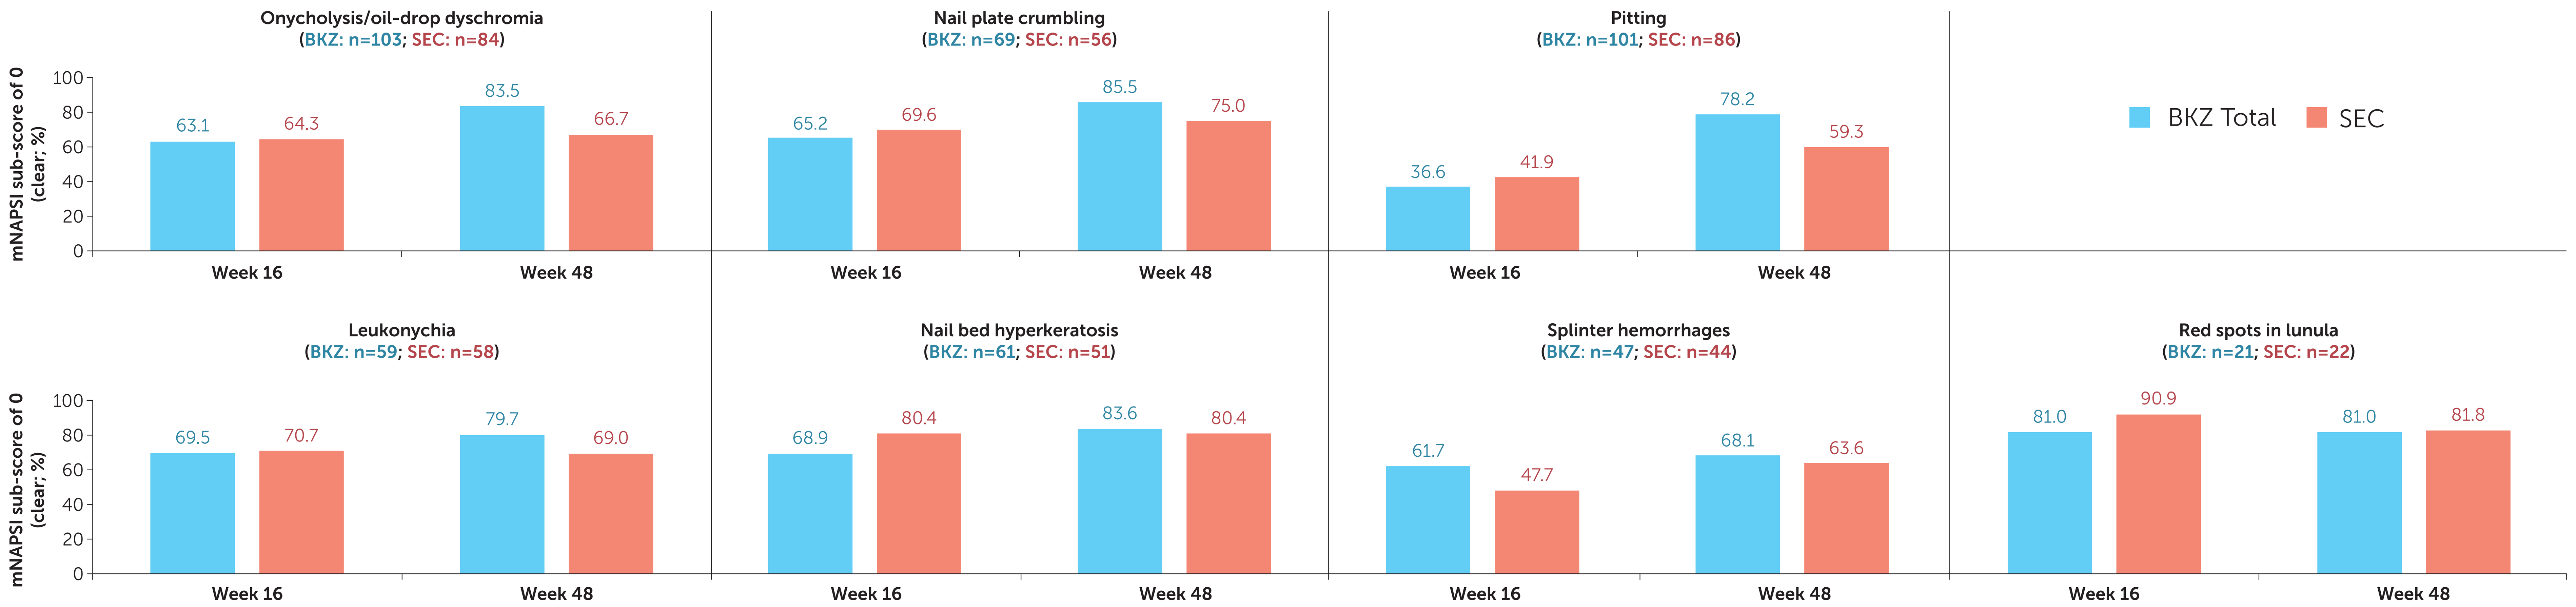
mNAPSI sub-score of 0 (clear; %)
Onycholysis/oil-drop dyschromia (BKZ: n=103; SEC: n=84)
Week 16: 63.1, 64.3
Week 48: 83.5, 66.7
Nail plate crumbling (BKZ: n=69; SEC: n=56)
Week 16: 65.2, 69.6
Week 48: 85.5, 75.0
Pitting (BKZ: n=101; SEC: n=86)
Week 16: 36.6, 41.9
Week 48: 78.2, 59.3
BKZ Total
SEC
Leukonychia (BKZ: n=59; SEC: n=58)
Week 16: 69.5, 70.7
Week 48: 79.7, 69.0
Nail bed hyperkeratosis (BKZ: n=61; SEC: n=51)
Week 16: 68.9, 80.4
Week 48: 83.6, 80.4
Splinter hemorrhages (BKZ: n=47; SEC: n=44)
Week 16: 61.7, 47.7
Week 48: 68.1, 63.6
Red spots in lunula (BKZ: n=21; SEC: n=22)
Week 16: 81.0, 90.9
Week 48: 81.0, 81.8

Only patients with mNAPSI >10 at baseline and who entered the maintenance period of BE RADIANT were included in this analysis. A score of 0 indicates clear nails.

BKZ: bimekizumab; **BSA:** body surface area; **DLQI:** Dermatology Life Quality Index; **IGA:** Investigator's Global Assessment; **mNAPSI:** modified Nail Psoriasis Severity Index; **mNRI:** modified non-responder imputation; **NAPSI:** Nail Psoriasis Severity Index; **NRI:** non-responder imputation; **OC:** observed case; **PASI:** Psoriasis Area and Severity Index; **Q4W:** every 4 weeks; **Q8W:** every 8 weeks; **SD:** standard deviation; **SEC:** secukinumab.

Institutions: [1]Department of Dermatology, Medical Center, University of Freiburg, Freiburg, Germany; [2]Department of Dermatology, The Icahn School of Medicine at Mount Sinai, New York, NY, USA; [3]Dermatology Unit, Department of Medicine, Università di Padova, Padova, Italy; [4]Department of Dermatology, University Hospital Carl Gustav Carus, TU Dresden, Dresden, Germany; [5]SKiN Centre for Dermatology, Probity Medical Research, Peterborough, ON, Canada, and Queen's University, Kingston, ON, Canada; [6]St Vincent's University Hospital, Elm Park and the Charles Institute, University College Dublin, Dublin, Republic of Ireland; [7]UCB Pharma, Slough, UK; [8]UCB Pharma, Morrisville, NC, USA; [9]UCB Pharma, Monheim, Germany; [10]UCB Pharma, Brussels, Belgium; [11]Department of Dermatology, UT Southwestern Medical Center, Dallas, TX, USA; [12]Department of Dermatology, University Hospitals of Cleveland, Case Western Reserve University, Cleveland, OH, USA.

References: [1]Augustin M. Br J Dermatol 2019;181:358–65; [2]Reich K. N Engl J Med 2021;385:142–52, NCT03536884; [3]Kaeley G. J Rheum 2021;48(8):1208–20.

Author Contributions: Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **KE**, **ABG**, **SP**, **SB**, **MG**, **BK**, **NT**, **CM**, **SW**, **DdC**, **JFM**, **BE**; Drafting of the publication, or reviewing it critically for important intellectual content: **KE**, **ABG**, **SP**, **SB**, **MG**, **BK**, **NT**, **CM**, **SW**, **DdC**, **JFM**, **BE**; Final approval of the publication: **KE**, **ABG**, **SP**, **SB**, **MG**, **BK**, **NT**, **CM**, **SW**, **DdC**, **JFM**, **BE**. **Author Disclosures: KE:** Speaker and/or advisor for AbbVie, Almirall, Boehringer Ingelheim, Bristol Myers Squibb, Eli Lilly, Janssen, LEO Pharma, Pfizer, Novartis, Sanofi, and UCB Pharma. **ABG:** Honoraria as an advisory board member and consultant for: Amgen, AnaptysBio, Avotres Therapeutics, Boehringer Ingelheim, Bristol Myers Squibb, DICE Therapeutics, Eli Lilly, Highlight Therapeutics, Janssen, Novartis, Sanofi, UCB Pharma, and Xbiotech; research/educational grants from: AnaptysBio, Bristol Myers Squibb, MoonLake Immunotherapeutics, Novartis, and UCB Pharma; all funds go to the Icahn School of Medicine at Mount Sinai. **SP:** Served as consultant and/or speaker for AbbVie, Almirall, Celgene, Eli Lilly, Janssen, LEO Pharma, Merck, Novartis, Pfizer, Sandoz, and UCB Pharma. **SB:** Honoraria from AbbVie, Actelion, Bristol Myers Squibb, Galderma, GSK, Janssen-Cilag, MSD, Novartis, and Roche Posay; honoraria for serving on advisory boards from AbbVie, Actelion, Amgen, Celgene, Eli Lilly, Galderma, Janssen-Cilag, LEO Pharma, Menlo Therapeutics, MSD, Novartis, Pfizer, and UCB Pharma. **MG:** Investigator, speaker, consultant or advisory board member for AbbVie, Akros, Amgen, AnaptysBio, Arcutis, Aslan, Bausch Health, Boehringer Ingelheim, Bristol Myers Squibb, Dermavant, Dermira, Eli Lilly, Galderma, GSK, Incyte, Janssen, Kyowa Kirin, MedImmune, Meiji, Merck, MoonLake Immunotherapeutics, Nimbus, Novartis, Pfizer, Regeneron, Reistone, Sanofi Genzyme, Sun Pharma, and UCB Pharma. **BK:** Received research support from or has been a principal investigator (clinical trials) for AbbVie, Almirall, Janssen, Merck, MoonLake Immunotherapeutics, Novartis, Pfizer, and UCB Pharma; has been a consultant for AbbVie, Almirall, Celgene, Eli Lilly, Janssen, Merck, MoonLake Immunotherapeutics, Novartis, Pfizer, and UCB Pharma; has received honoraria from AbbVie, Almirall, Celgene, Janssen, Eli Lilly, MoonLake Immunotherapeutics, Novartis, Pfizer, and UCB Pharma; and has been on scientific advisory boards for AbbVie, Almirall, Celgene, Janssen, Eli Lilly, MoonLake Immunotherapeutics, Novartis, Pfizer, and UCB Pharma. **NT**, **CM**, **SW**, **DdC:** Employees and shareholders of UCB Pharma. **JFM:** Consultant and/or investigator for AbbVie, Amgen, Biogen, Bristol Myers Squibb, Dermavant, Eli Lilly, Janssen, LEO Pharma, Pfizer, Novartis, Regeneron, Sanofi, Sun Pharma, and UCB Pharma. **BE:** Received research support as funding to Case Western Reserve University from AbbVie, AnaptysBio, Boehringer Ingelheim, Bristol Myers Squibb, Celgene, Eli Lilly, Incyte, LEO Pharma, Menlo, Merck, Novartis, Pfizer, Regeneron, Sun Pharma, Valeant, and Vanda; consultant (honoraria) for Arcutis, Boehringer Ingelheim, Bristol Myers Squibb, Celgene, Eli Lilly, LEO Pharma, Menlo, Novartis, Pfizer, Sun Pharma, UCB Pharma, Valeant, and Verrica. **Acknowledgements:** We would like to thank the patients and their caregivers in addition to all the investigators and their teams who contributed to this study. The authors acknowledge Joe Dixon, PhD, UCB Pharma, Slough, UK for publication coordination, Jack Wardle, MSc, Costello Medical, Cambridge, UK, for medical writing and editorial assistance and the Creative team at Costello Medical for graphic design assistance. This study was funded by UCB Pharma. All costs associated with development of this presentation were funded by UCB Pharma.

To receive a copy of this poster, scan the QR code or visit: UCBposters.com/ACR2023

Poster ID: 2255
Link expiration: November 29, 2023

Presented at ACR Convergence 2023 | November 10–15 | San Diego, CA

Previously presented at AAD 2023 | March 17–21 | New Orleans, LA